LUNG PARTITIONING OF ΔF508-CFTR CORRECTORS

John Ferkany, Mauri Krouse, Andrew Kolodziej, Richard Fitzpatrick, Bridget Cole
Flatley Discovery Lab, LLC. 529 Main Street, Charlestown, MA 02129

INTRODUCTION: The CF community has made substantial progress advancing promising new therapies into clinical trials to treat the underlying cause of this devastating genetic disease. As investigational drugs and combinations thereof progress in clinical development, it is increasingly evident that the path to a cure will require a combination treatment of two or more drugs. This poses additional challenges managing potential drug-drug interactions. One strategy to reduce the risk of drug-drug interactions is to develop a molecule that partitions well into lung tissue while having low drug levels in systemic circulation. We describe the lung to plasma ratio of ΔF508-CFTR correctors following intravenous and oral dosing to SD rats.

METHODS AND RESULTS:
Sprague Dawley rats (n= 5) were injected intravenously with a solution containing CFTR correctors FDL169 and VX809 at a dose of 1 mg/kg each. Plasma and lung samples were collected at 6 time points over 12 hours post administration. Exposure of FDL169 and VX809 were determined in lung and plasma samples, and the data were used to estimate a lung to plasma ratio at each time point. In this experiment, the lung to plasma ratio for FDL169 and VX809 ranged from 2.2 to 3.7 and 0.37 to 0.5, respectively.

Distribution of Detected Drug Following IV Cassette Dosing of FDL169 and VX809

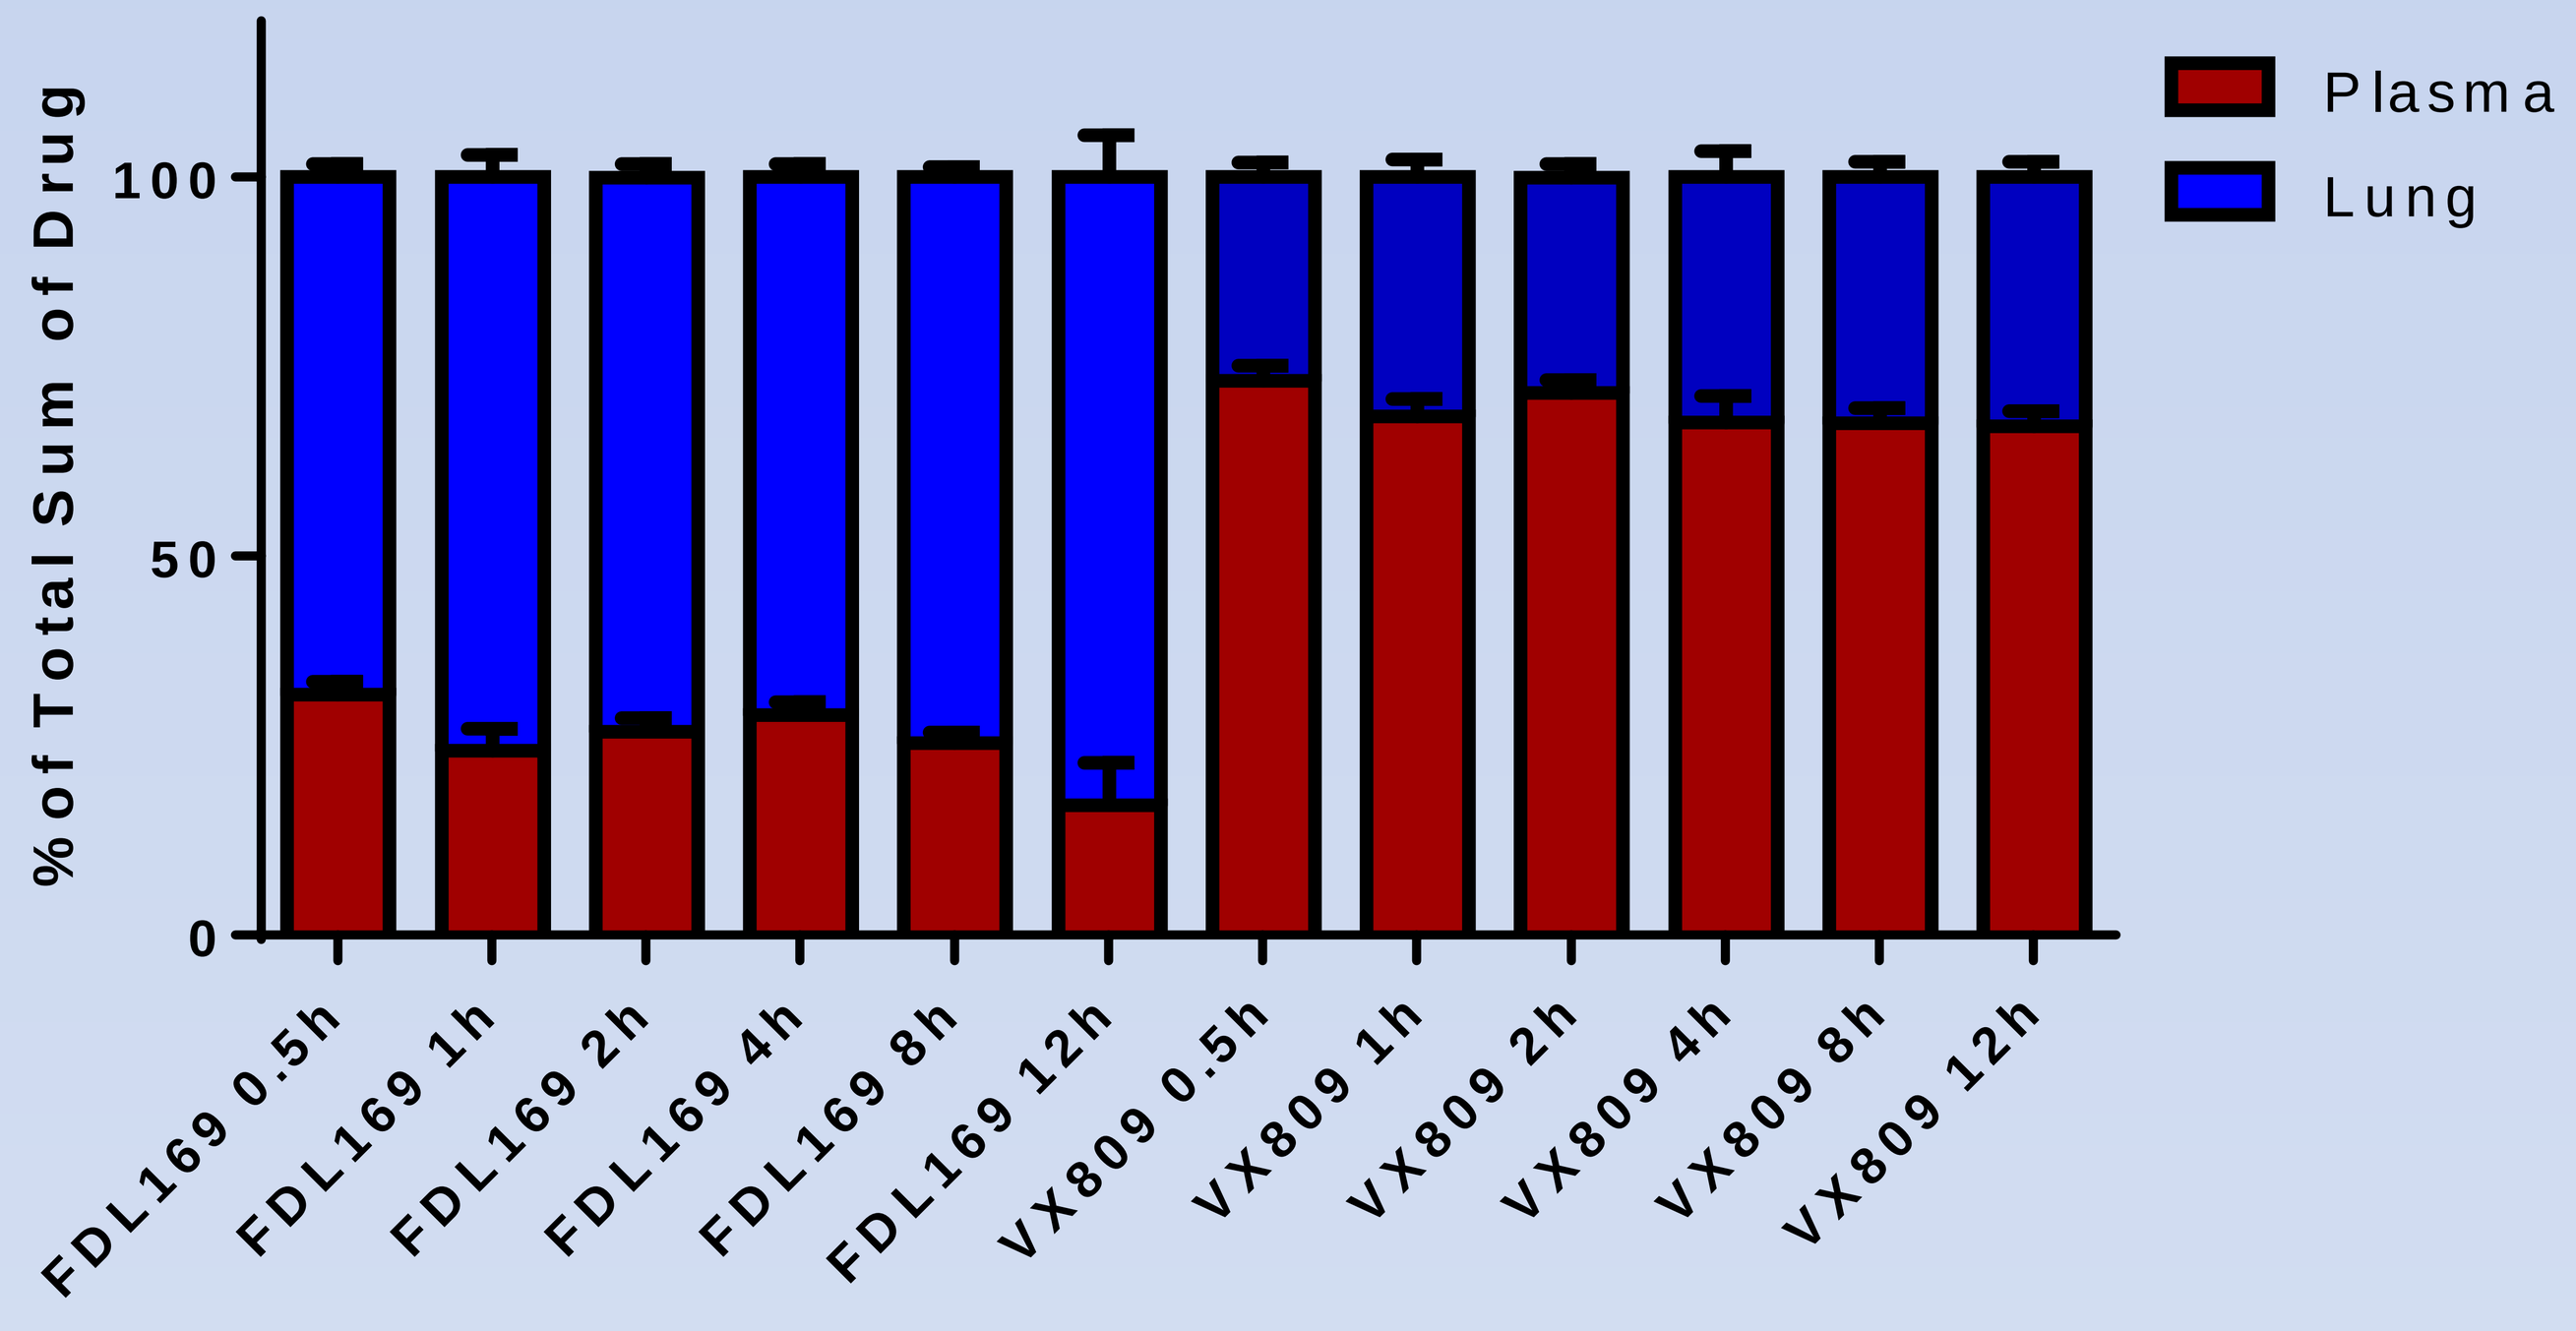
% of Total Sum of Drug
0
50
100
Plasma
Lung
FDL169 0.5h
FDL169 1h
FDL169 2h
FDL169 4h
FDL169 8h
FDL169 12h
VX809 0.5h
VX809 1h
VX809 2h
VX809 4h
VX809 8h
VX809 12h

Lung/Plasma Ratios in SD Rats

Time	FDL169			VX809		
(h)	Mean	SD	%CV	Mean	SD	%CV
0.5	2.2	0.4	17.3%	0.37	0.1	22.5%
1	3.4	1.4	42.0%	0.47	0.1	23.4%
2	2.8	0.6	20.7%	0.40	0.1	17.8%
4	2.5	0.5	20.4%	0.50	0.2	36.6%
8	3.0	0.4	14.8%	0.49	0.1	22.4%
12	3.7	1.2	32.7%	0.50	0.1	20.7%

In a subsequent study, FDL169 was dosed orally at 30 mg/kg in SD rats. Test article FDL169 was administered as a solution in 1:3 NMP/PEG 400 vehicle. Plasma and lung samples were collected over 24 hours and the exposure of FDL169 was determined in each sample. The exposure data were used to estimate a lung to plasma ratio at each time point. The lung to plasma ratio for FDL169 in this experiment ranged from 1.6 at 30 min to 2.7 at 24 hours post administration.

Lung and Plasma Exposure of FDL169 Following a Single Oral Dose of 30 mg/kg in SD Rats

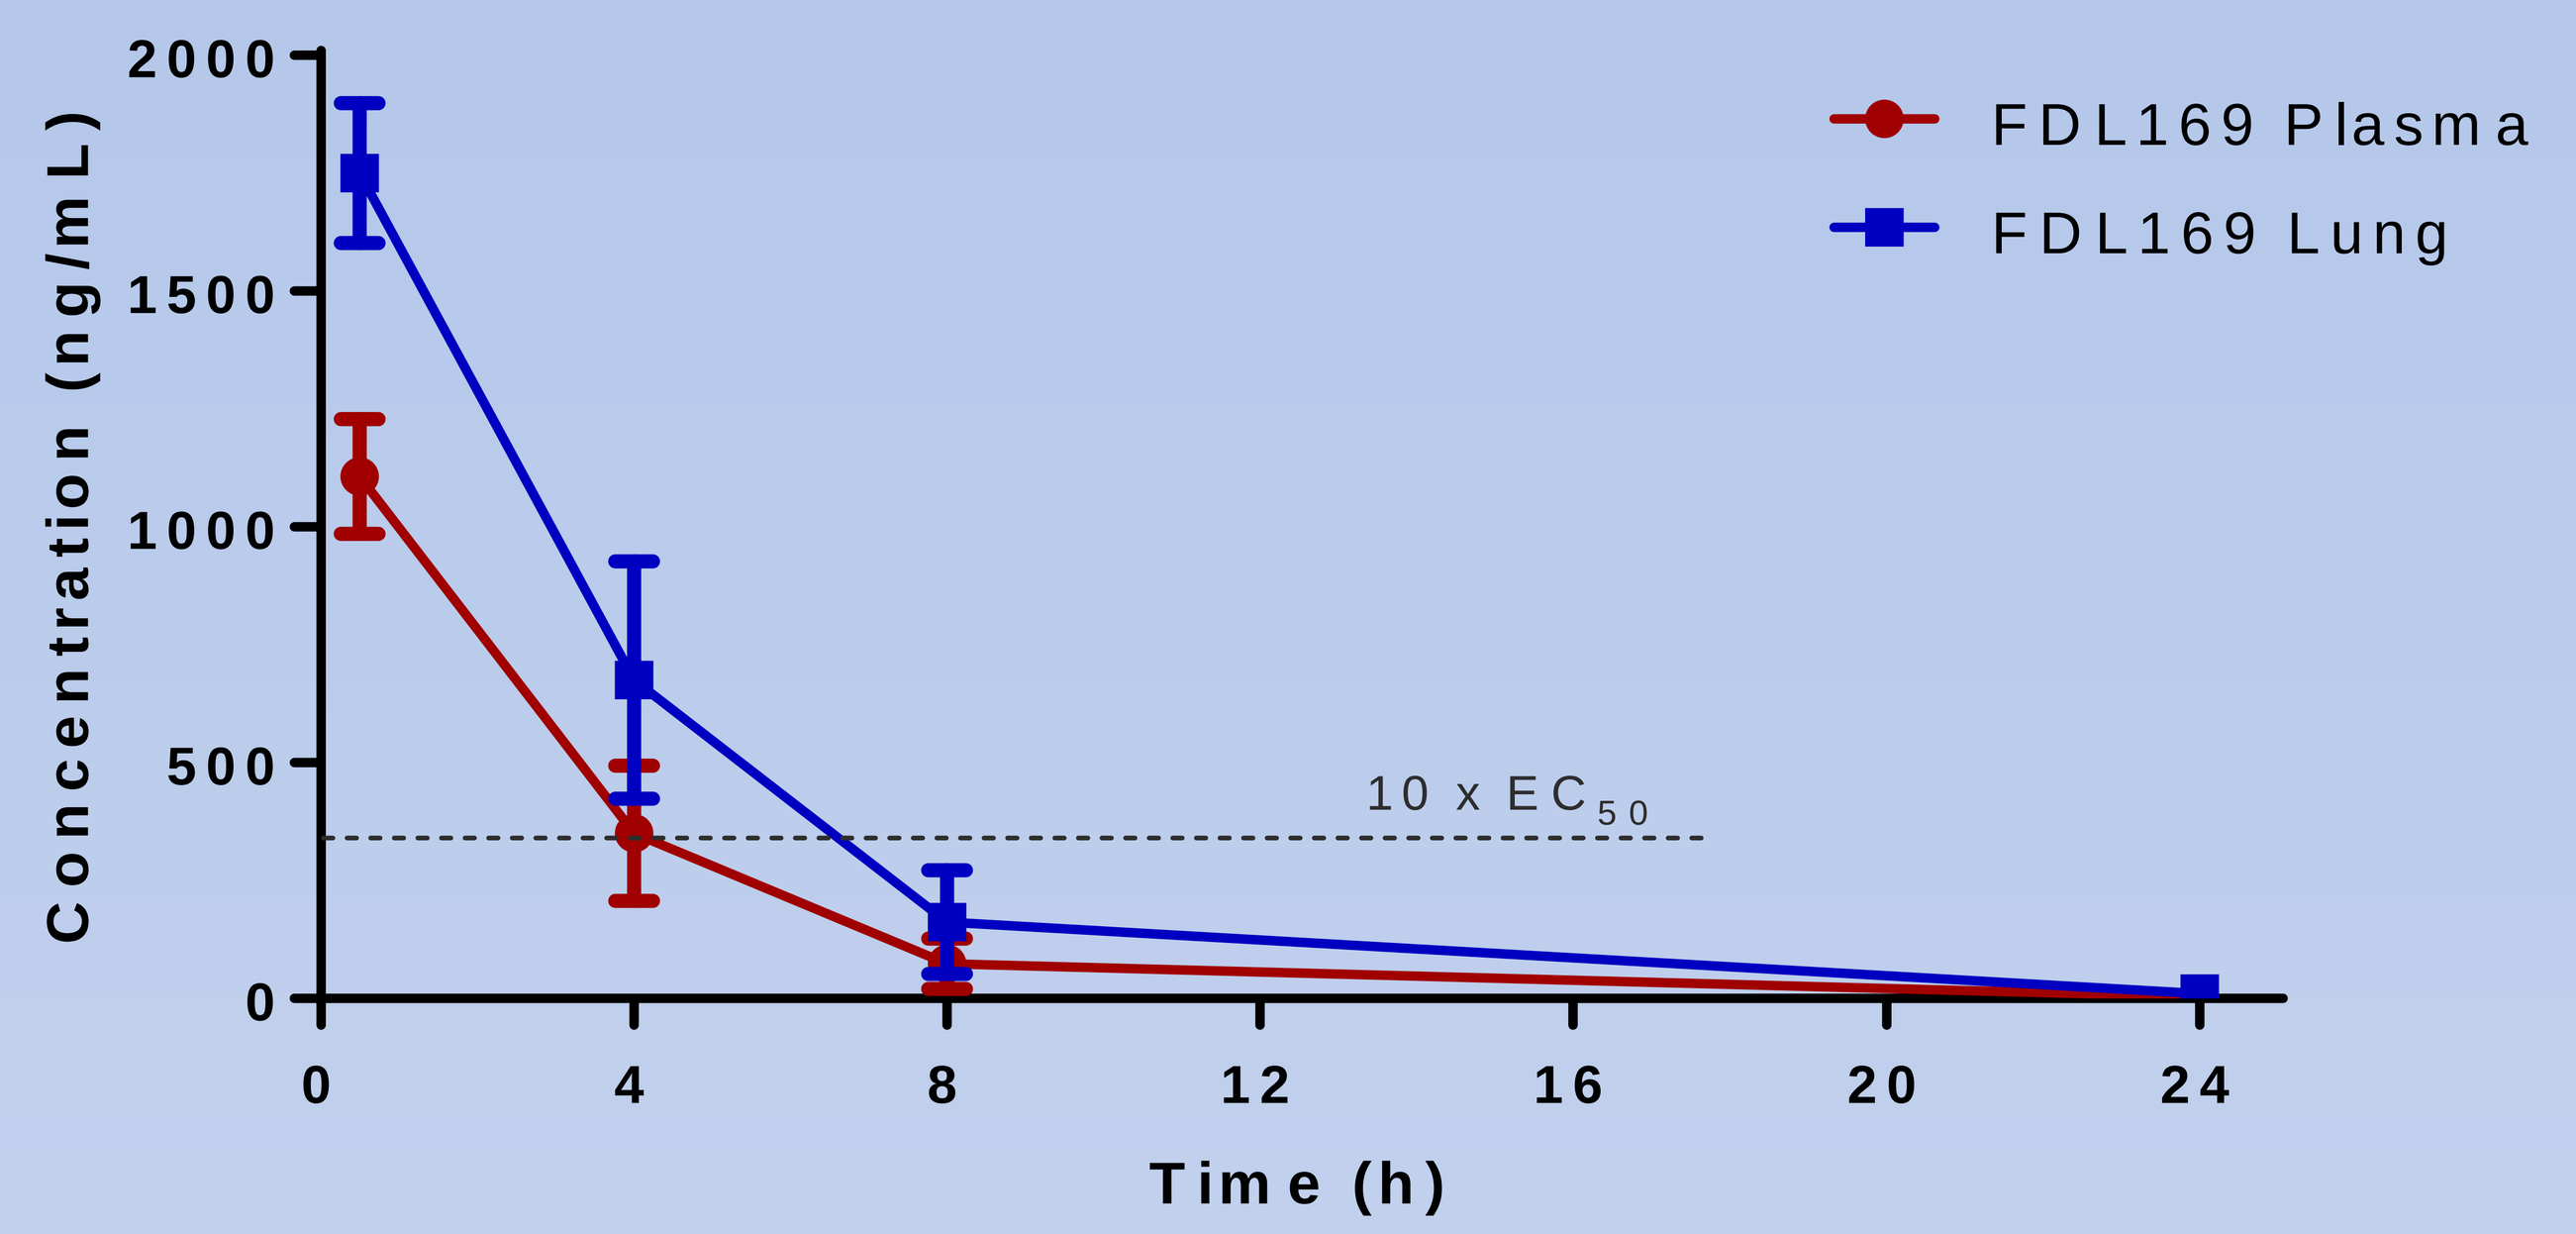
Concentration (ng/mL)
0
500
1000
1500
2000
FDL169 Plasma
FDL169 Lung
10 x EC50
0
4
8
12
16
20
24
Time (h)

Lung/Plasma Ratios

Time	Animal						
(h)	1	2	3	4	Mean	SD	%CV
0.5	1.4	1.5	2.0	1.5	**1.6**	0.3	17.8%
4	2.4	1.8	2.1	1.8	**2.0**	0.3	13.7%
8	2.4	2.4	2.1	2.5	**2.4**	0.2	7.2%
24	2.6	2.0	3.0	3.2	**2.7**	0.5	19.6%

In order to estimate the FDL169 lung/plasma ratio at steady state FDL169 was dosed once daily at 52 mg/kg in SD rats for 7 days. Test article was administered as an amorphous aqueous dispersion via oral gavage. Plasma and lung samples were collected on day 1 at 4, 8 and 24 hours, and day 7 at 4, 8, 24, 32 and 48 hours. Samples were analyzed for FDL169 exposure. The lung to plasma ratio for FDL169 on day 1 and day 7 were essentially similar. By day 7 FDL169 lung/plasma ratios ranged from 2.1 at 4 hours to 18.9 at 48 hours post administration.

Lung to Plasma Ratio

Time (h)	Animal						
Day 1	**1**	**2**	**3**	**4**	**Mean**	**SD**	**%CV**
4 h	3.6	3.3	3.1	2.8	**3.2**	0.3	10.13%
8 h	4.2	3.2	4.0	3.7	**3.8**	0.4	11.38%
24 h	4.7	2.3	2.6	4.3	**3.5**	1.2	35.11%
Day 7	**1**	**2**	**3**	**4**	**Mean**	**SD**	**%CV**
4 h	2.2	2.0	1.9	2.3	**2.1**	0.2	9.96%
8 h	3.8	3.2	3.6	4.3	**3.7**	0.5	12.72%
24 h	3.8	4.8	4.3	4.2	**4.3**	0.4	9.16%
32 h	5.2	5.3	5.8	0.8	**4.3**	2.3	54.40%
48 h	3.8	14.0	35.4	22.4	**18.9**	13.4	70.81%

Lung and Plasma Exposure of FDL169 Following a 7-Day Repeat Dosing in SD Rats

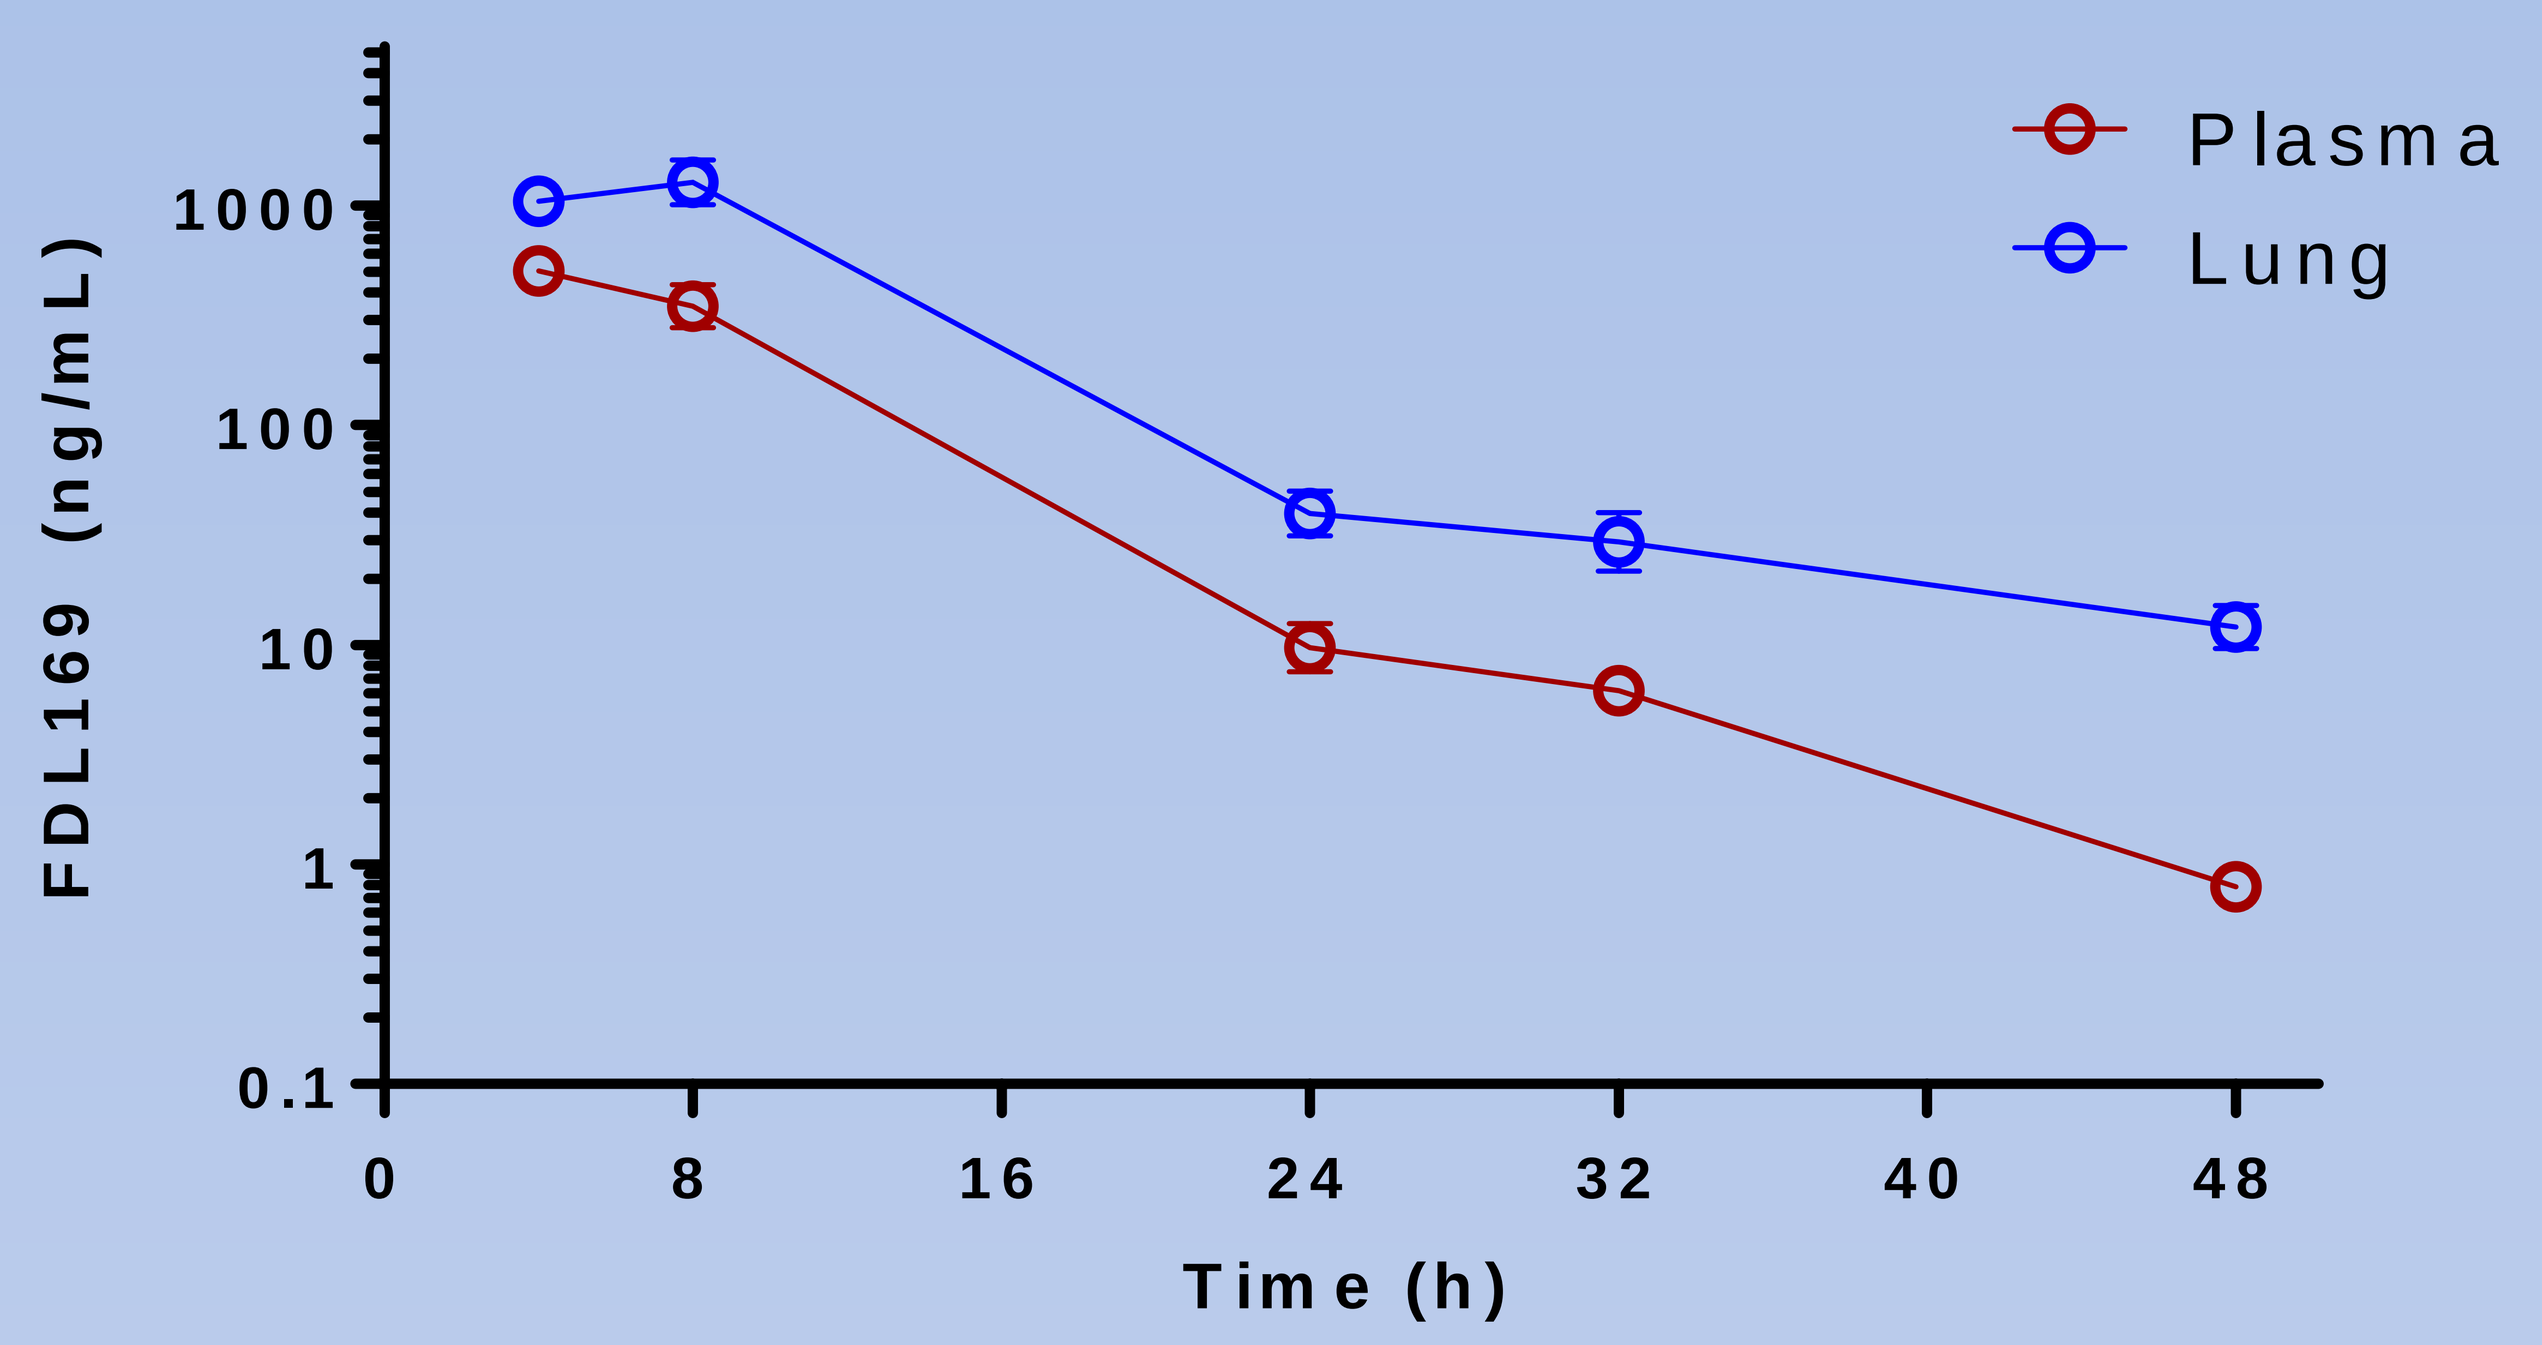
FDL169 (ng/mL)
0.1
1
10
100
1000
Plasma
Lung
0
8
16
24
32
40
48
Time (h)

SUMMARY OF RESULTS:

- In general, data from the experiments presented demonstrate that FDL169 exposure is ≥ 2-fold higher in the lung than systemic circulation following IV and oral administration in SD rats.
- VX809 demonstrates ≥ 2-fold lower drug levels in the lung than systemic circulation following IV administration in SD rats.
- The lung/plasma ratio of FDL169 ranged from 2.2 to 3.7 following IV cassette dosing
- The lung/plasma ratio of VX809 ranged from 0.37 to 0.5 following IV cassette dosing
- FDL169 lung/plasma ratio ranged from 1.6 at 30 min to 2.7 at 24 hours following single oral dose.
- FDL169 lung/plasma ratios ranged from 2.1 at 4 hours to 18.9 at 48 hours following once daily dosing at 52 mg/kg in SD rats for 7 days.
- FDL169 lung/plasma ratios are essentially similar on day 1 and day 7.
- FDL169 and VX809 were well tolerated with no adverse events observed in study animals from all dose groups.

CONCLUSION: Correctors FDL169 and VX809 demonstrate comparable *in vitro* potency and efficacy in primary ΔF508-CFTR human bronchial epithelial cells. However, the *in vivo* properties of these two clinical candidates are markedly different. FDL169 partitions well into lung tissue with exposure levels ≥ 2-fold higher (≥ 200%) than systemic circulating drug levels. In contrast, the lung exposure of VX809 is approximately 40% of systemic circulating concentrations. These data suggest that FDL169 has superior distribution to lung tissue compared to VX809 and should therefore require lower levels of drug in systemic circulation to achieve lung concentrations that promote clinical efficacy. Reduced systemic exposure of FDL169 is expected to lower the risk of drug-drug interactions and off-target events in patients.

Flatley Discovery Lab Seeking A Cure for Cystic Fibrosis