Distinguishing Properties of CFTR Potentiator FDL176

P. Bhatt, W. An, V. Bailey, C. Bresilla, J.Chin, I. Kwok, B. Cole, M.E. Krouse

Flatley Discovery Lab, Charlestown, MA United States

Summary: FDL176 is a novel CFTR potentiator that stimulates chloride current in F508del-CFTR primary CF airway epithelial cells with similar *in vitro* efficacy to ivacaftor and an EC_{50} of 127 nM. In FRT cells expressing F508del-CFTR, stimulation with FDL176 increases chloride current >2-fold suggesting the open probability of uncorrected F508del-CFTR increases at least 2-fold upon acute exposure. The activity of FDL176 and ivacaftor following acute treatment was compared in other CF causing mutations expressed in FRT cells. In G551D cells, FDL176 increases chloride current by 10-fold and ivacaftor by 13-fold. In N1303K cells, potentiation with FDL176 and Ivacaftor increase chloride current by 3-fold and 4-fold, respectively. Both potentiators increase chloride current by 2-fold in R117H cells and 1.2 to 1.3-fold was observed for mutations G85E, E92K and R560T. The potency of both potentiators was similar in mutations R560T, G85E and EK92; potency of FDL176 was ≥ 10-fold higher than ivacaftor in mutations R117H, F508del, G551D and N1303K. In tezacaftor corrected F508del-CFTR primary cells, chronic exposure to ivacaftor (≥ 6 hours) reduced chloride current by >50% compared to acute conditions (0 hrs). In FDL169 corrected cells chronic exposure to FDL176 reduced chloride current by <20% whereas chronic exposure to ivacaftor reduce chloride current by >30%. The reduction of chloride current does not change in presence of human serum, but the IC_{50} of both potentiators shifts to higher concentrations. HRP cell surface expression in CFBE cells is not affected by acute exposure to FDL176 or ivacaftor, but prolonged exposure reduces the amount of CFTR at the cell surface by 36% for FDL176 and 57% for ivacaftor. In summary, FDL176 and ivacaftor produce a similar *in vitro* response upon acute stimulation, while the impact of chronic exposure to FDL176 on chloride current is less than ivacaftor.

Figure 1: FDL176 has similar efficacy and potency in FDL169, tezacaftor and lumacaftor corrected F508del-CFTR hBE cells

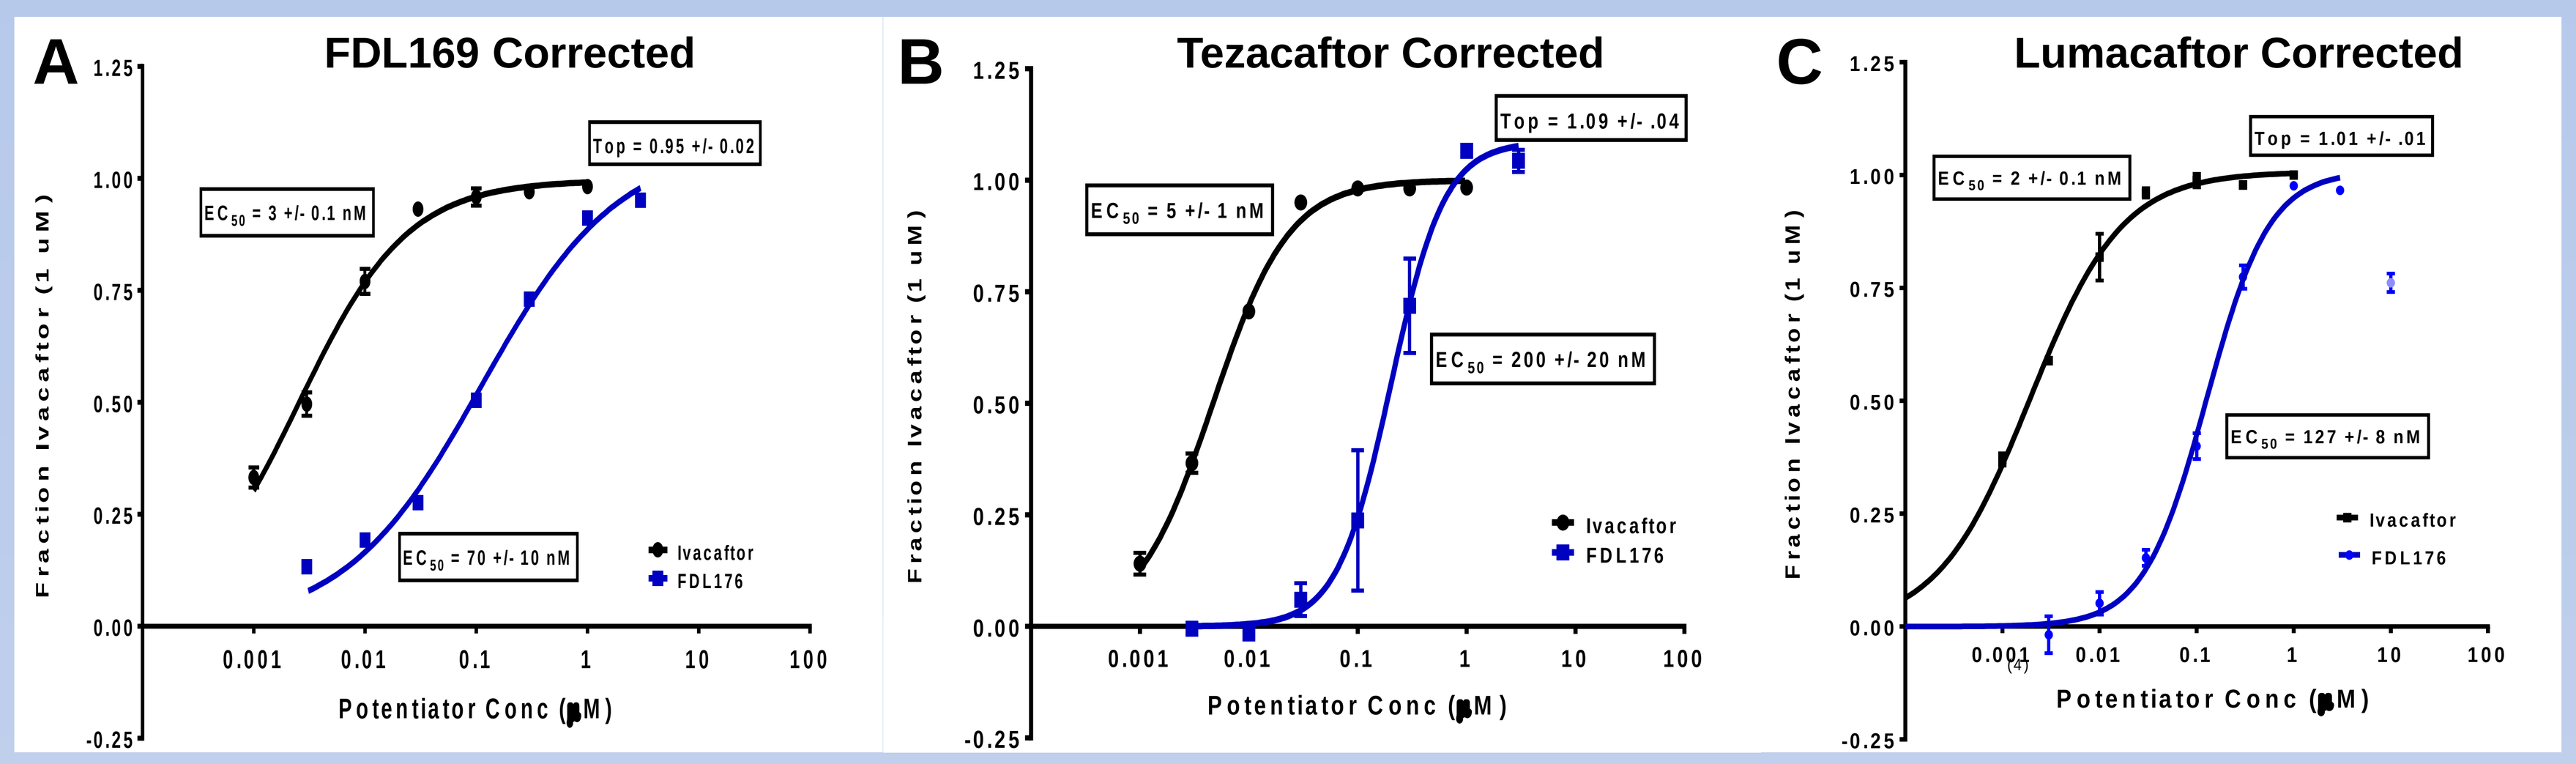
A FDL169 Corrected
EC_{50} = 3 +/- 0.1 nM
Top = 0.95 +/- 0.02
EC_{50} = 70 +/- 10 nM
B Tezacaftor Corrected
EC_{50} = 5 +/- 1 nM
Top = 1.09 +/- .04
EC_{50} = 200 +/- 20 nM
C Lumacaftor Corrected
EC_{50} = 2 +/- 0.1 nM
Top = 1.01 +/- .01
EC_{50} = 127 +/- 8 nM
Ivacaftor
FDL176
Fraction Ivacaftor (1 uM)
Potentiator Conc (µM)

- Primary CFhBE cells treated with **(A)** FDL169 , **(B)** Tezacaftor and **(C)** Lumacaftor
- Acute stimulation with potentiator + forskolin
- Chloride transport measured using the TECC-24 equivalent current assay
- Maximum chloride transport response of FDL176 vs Ivacaftor is similar
- FDL176 is less potent than ivacaftor with EC_{50} ranging from 70-200 nM

Figure 2: Chloride Transport is higher in F508del-CFTR hBE cells exposed to FDL176 compared to ivacaftor under chronic treatment conditions

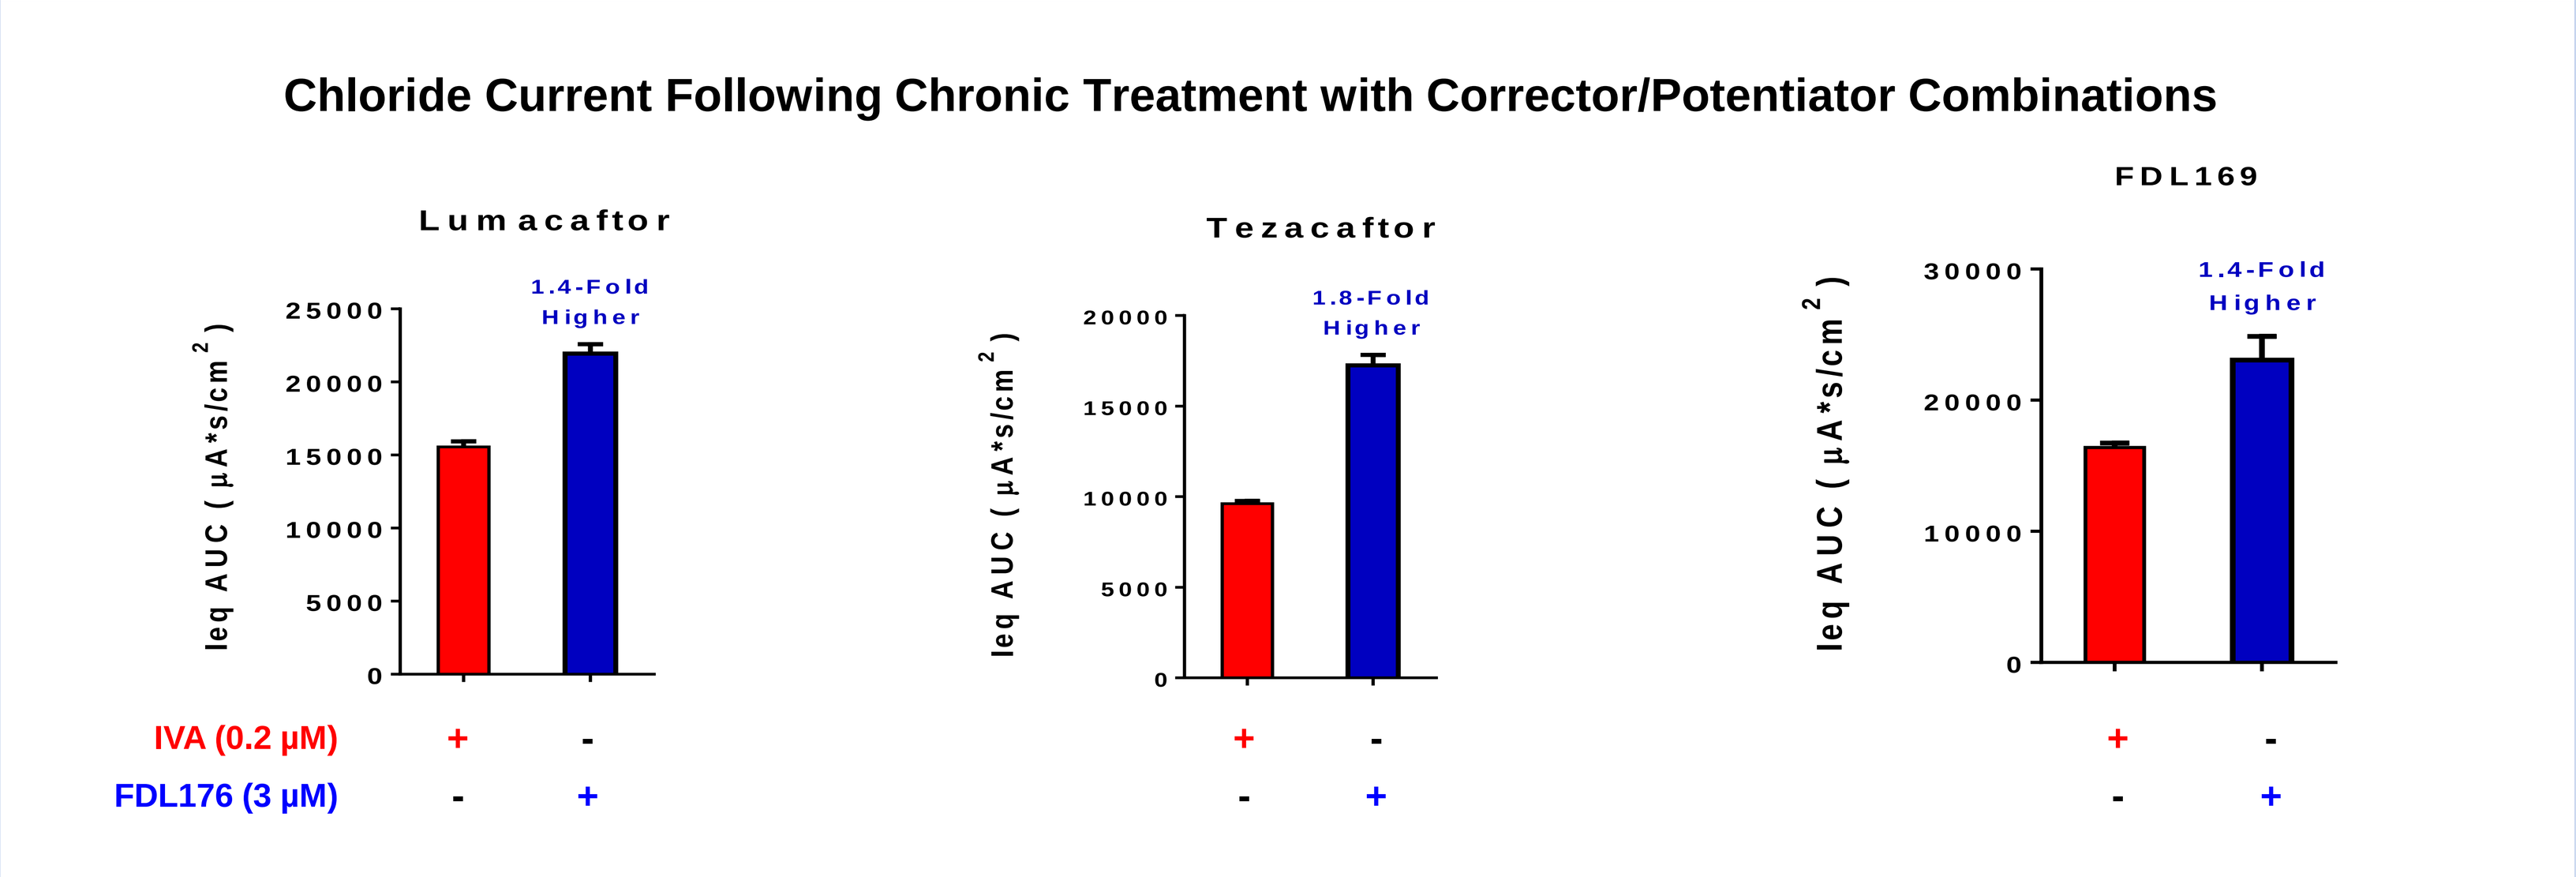
Chloride Current Following Chronic Treatment with Corrector/Potentiator Combinations
Lumacaftor
1.4-Fold Higher
Tezacaftor
1.8-Fold Higher
FDL169
1.4-Fold Higher
Ieq AUC (µA*s/cm²)
IVA (0.2 µM)
FDL176 (3 µM)

- Primary F508del-CFTR hBE cells were incubated with potentiator + corrector combinations for 24 hours and acutely stimulated with forskolin + 1 µM ivacaftor
- Chloride transport measured using the TECC-24 equivalent current assay
- Maximum chloride transport response of FDL176 is higher than ivacaftor

Figure 3: FDL176 achieves higher F508del-CFTR cell surface expression than ivacaftor in corrected CFBE41o- cells under chronic conditions

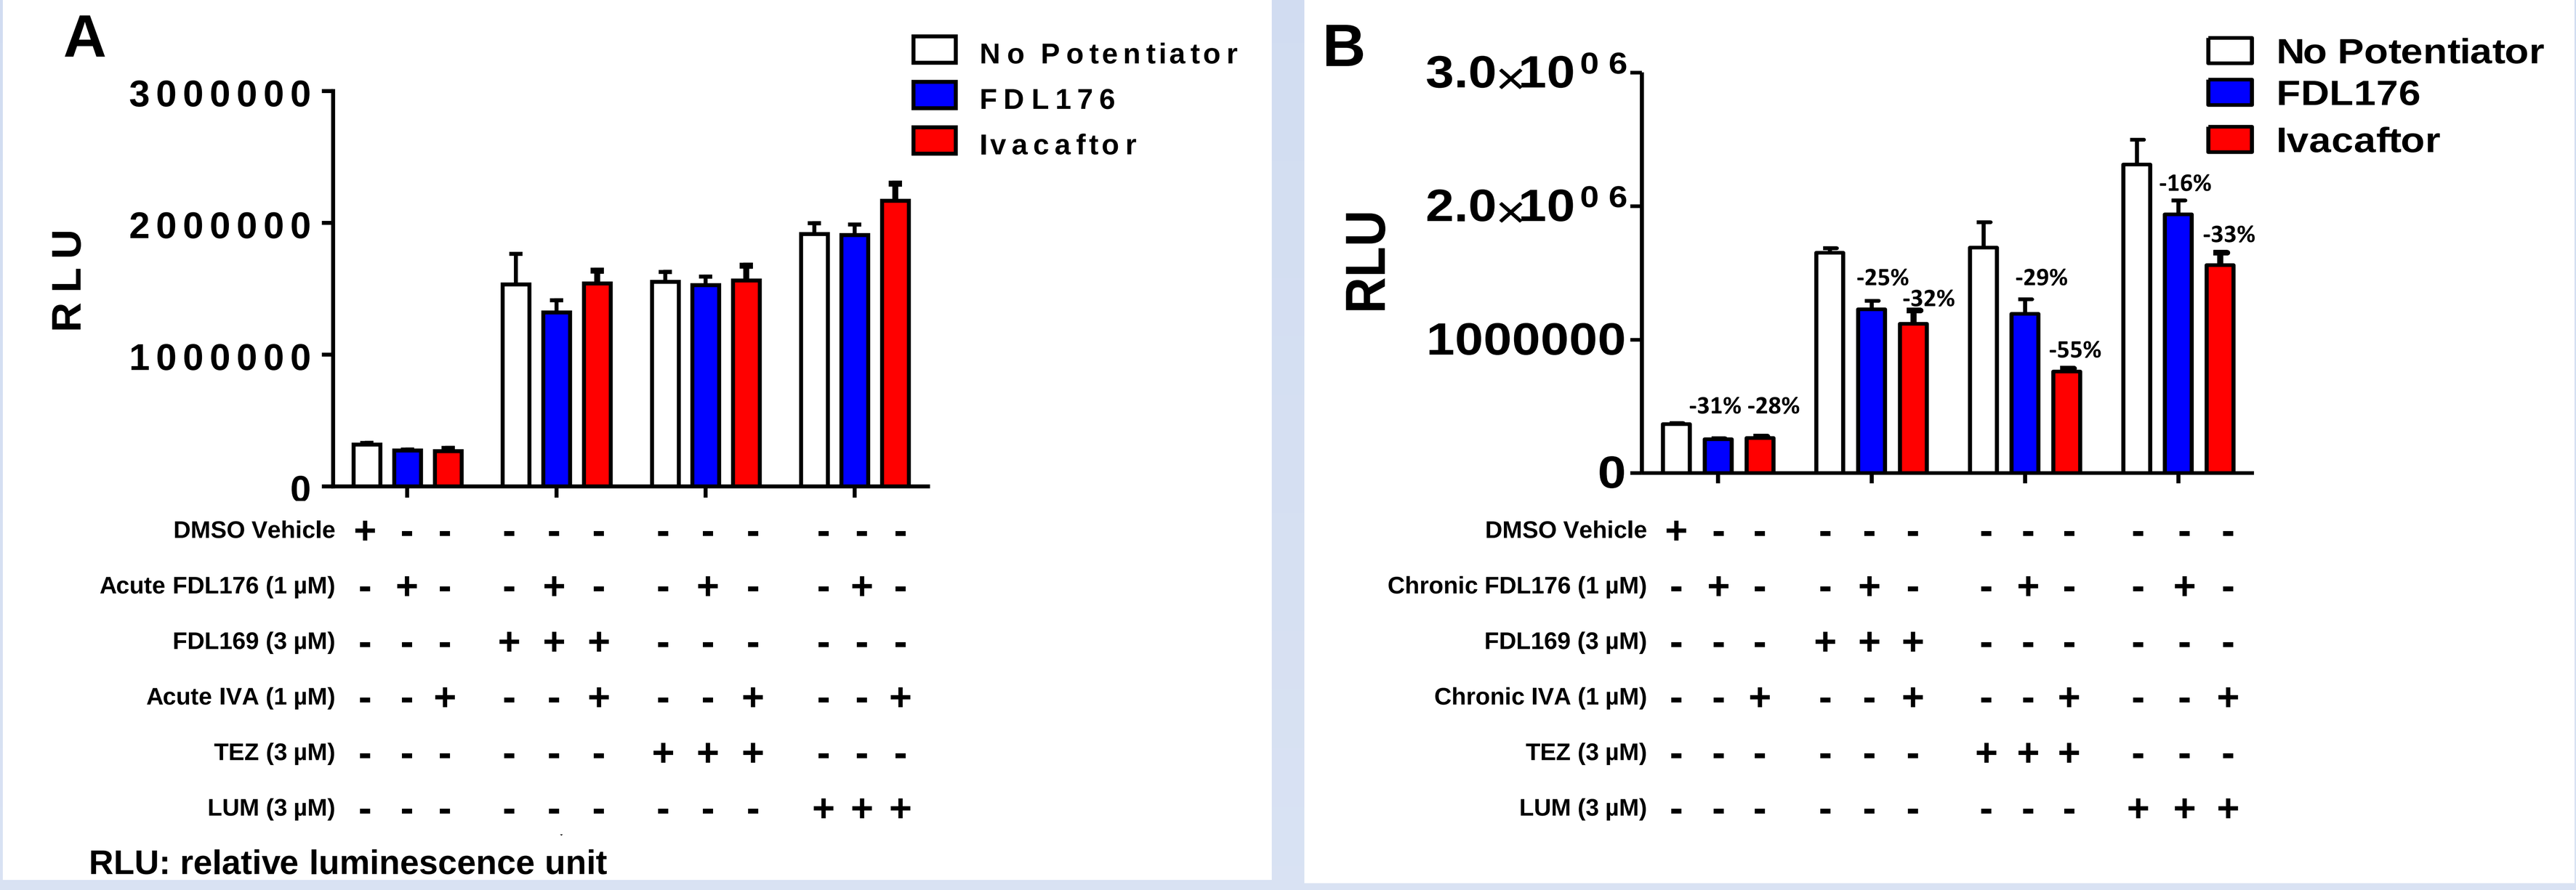
A
No Potentiator
FDL176
Ivacaftor
RLU
DMSO Vehicle
Acute FDL176 (1 µM)
FDL169 (3 µM)
Acute IVA (1 µM)
TEZ (3 µM)
LUM (3 µM)
RLU: relative luminescence unit
B
-31% -28%
-25%
-32%
-29%
-55%
-16%
-33%
Chronic FDL176 (1 µM)
Chronic IVA (1 µM)

- HRP cell surface assay in CFBE41o- cells
- Cells treated with corrector FDL169, tezacaftor or lumacaftor for 24 hours
- Figure A: Potentiators added acutely do not effect CFTR expression compared to control
- Figure B:Chronic treatment (24 hour) with corrector + potentiator reduced surface expression of F508del CFTR compared to control (no potentiator)
- The largest reduction of F508del-CFTR expression observed (-55%) was from the tezacaftor + ivacaftor combination

Figure 4: Human Serum Does Not Reduce Chronic Inhibition by Potentiators

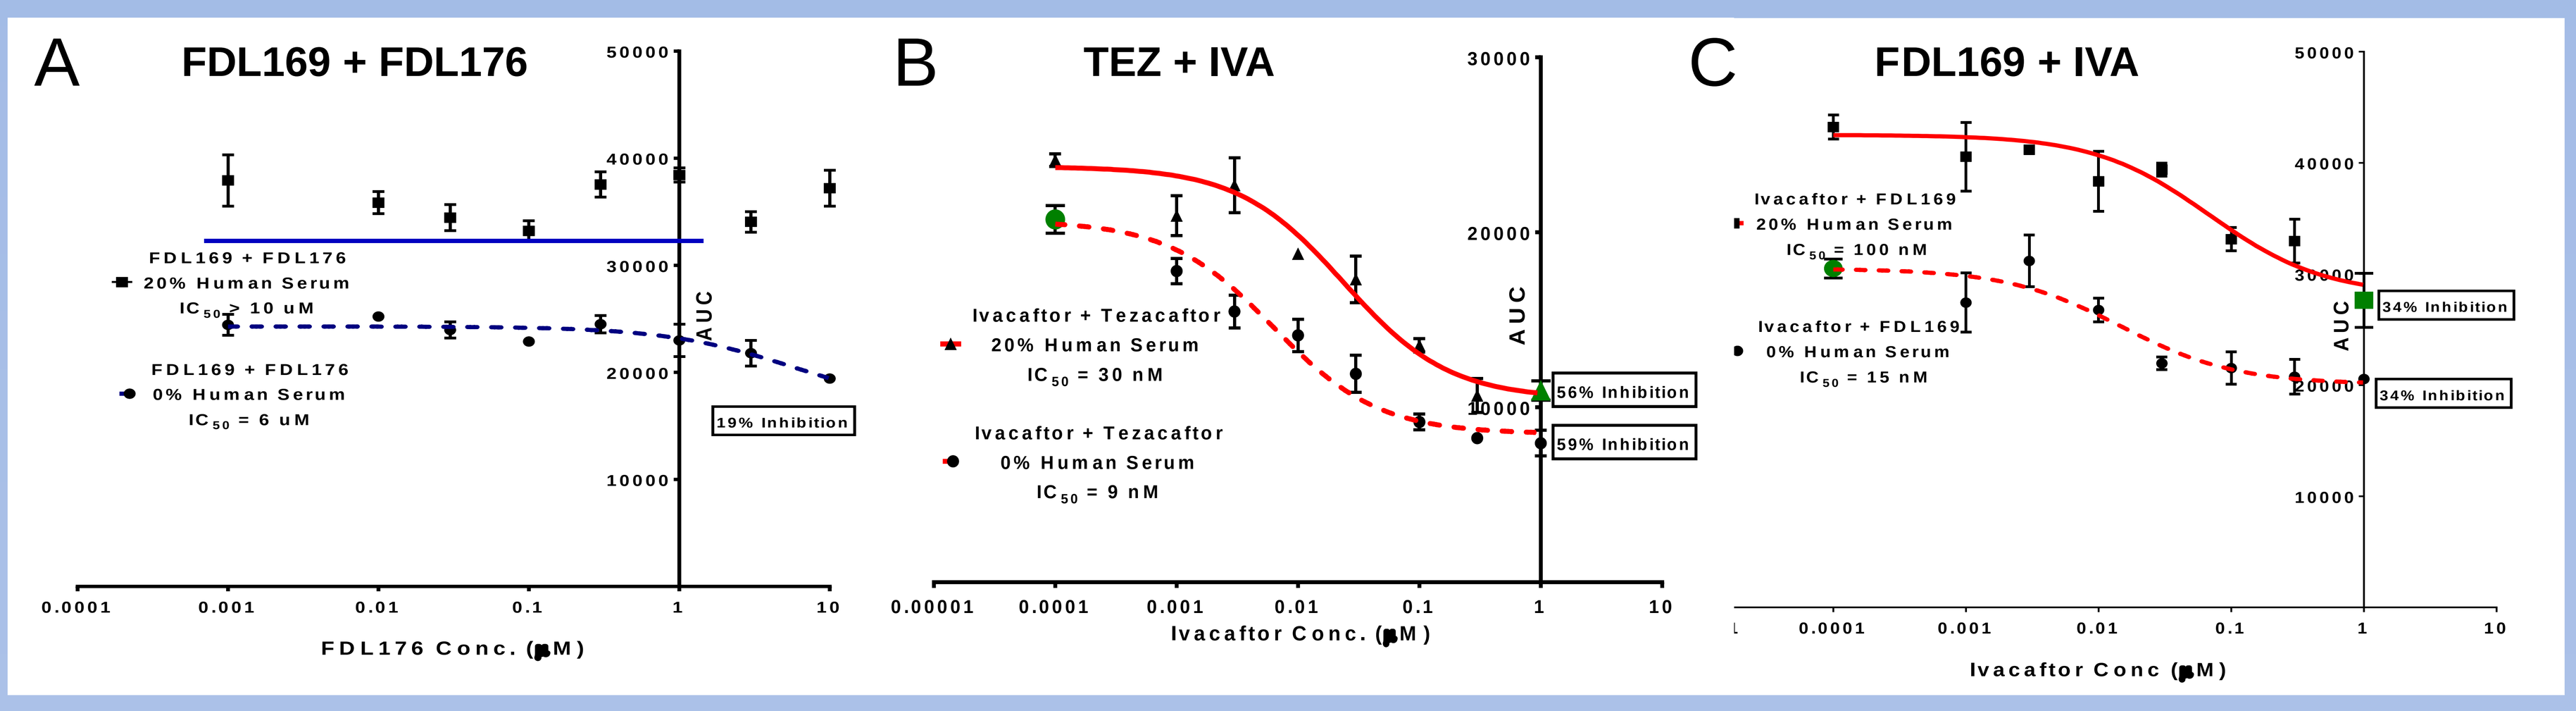
A FDL169 + FDL176
FDL169 + FDL176 20% Human Serum IC_{50} > 10 uM
FDL169 + FDL176 0% Human Serum IC_{50} = 6 uM
19% Inhibition
FDL176 Conc. (µM)
B TEZ + IVA
Ivacaftor + Tezacaftor 20% Human Serum IC_{50} = 30 nM
Ivacaftor + Tezacaftor 0% Human Serum IC_{50} = 9 nM
56% Inhibition
59% Inhibition
Ivacaftor Conc. (µM)
C FDL169 + IVA
Ivacaftor + FDL169 20% Human Serum IC_{50} = 100 nM
Ivacaftor + FDL169 0% Human Serum IC_{50} = 15 nM
34% Inhibition
34% Inhibition
Ivacaftor Conc (µM)
AUC

- Primary CFhBE cells were incubated with test compounds ± 20% human serum for 24 hours and subsequently stimulated with forskolin + 1uM ivacaftor
- The addition of human serum 24 hours before the experiment increases the CFTR current. If one uses the incorrect negative control, it may appear there is no chronic inhibition by potentiation. (compare green dots in Figure C)
- Figure A: FDL176 + FDL169; The lack of chronic inhibition seen with 20% human serum is most likely due to a shift in the IC_{50} to a concentration >10 uM
- Figure B: tezacaftor + ivacaftor treated cells show ~58% inhibition regardless of the addition of human serum
- Figure C: FDL169 + ivacaftor treated cells show less inhibition than tezacaftor + ivacaftor cells, but the amount inhibition is similar ± serum

Figure 5: FDL176 Increases Open Probability of F508del-CFTR >2-fold

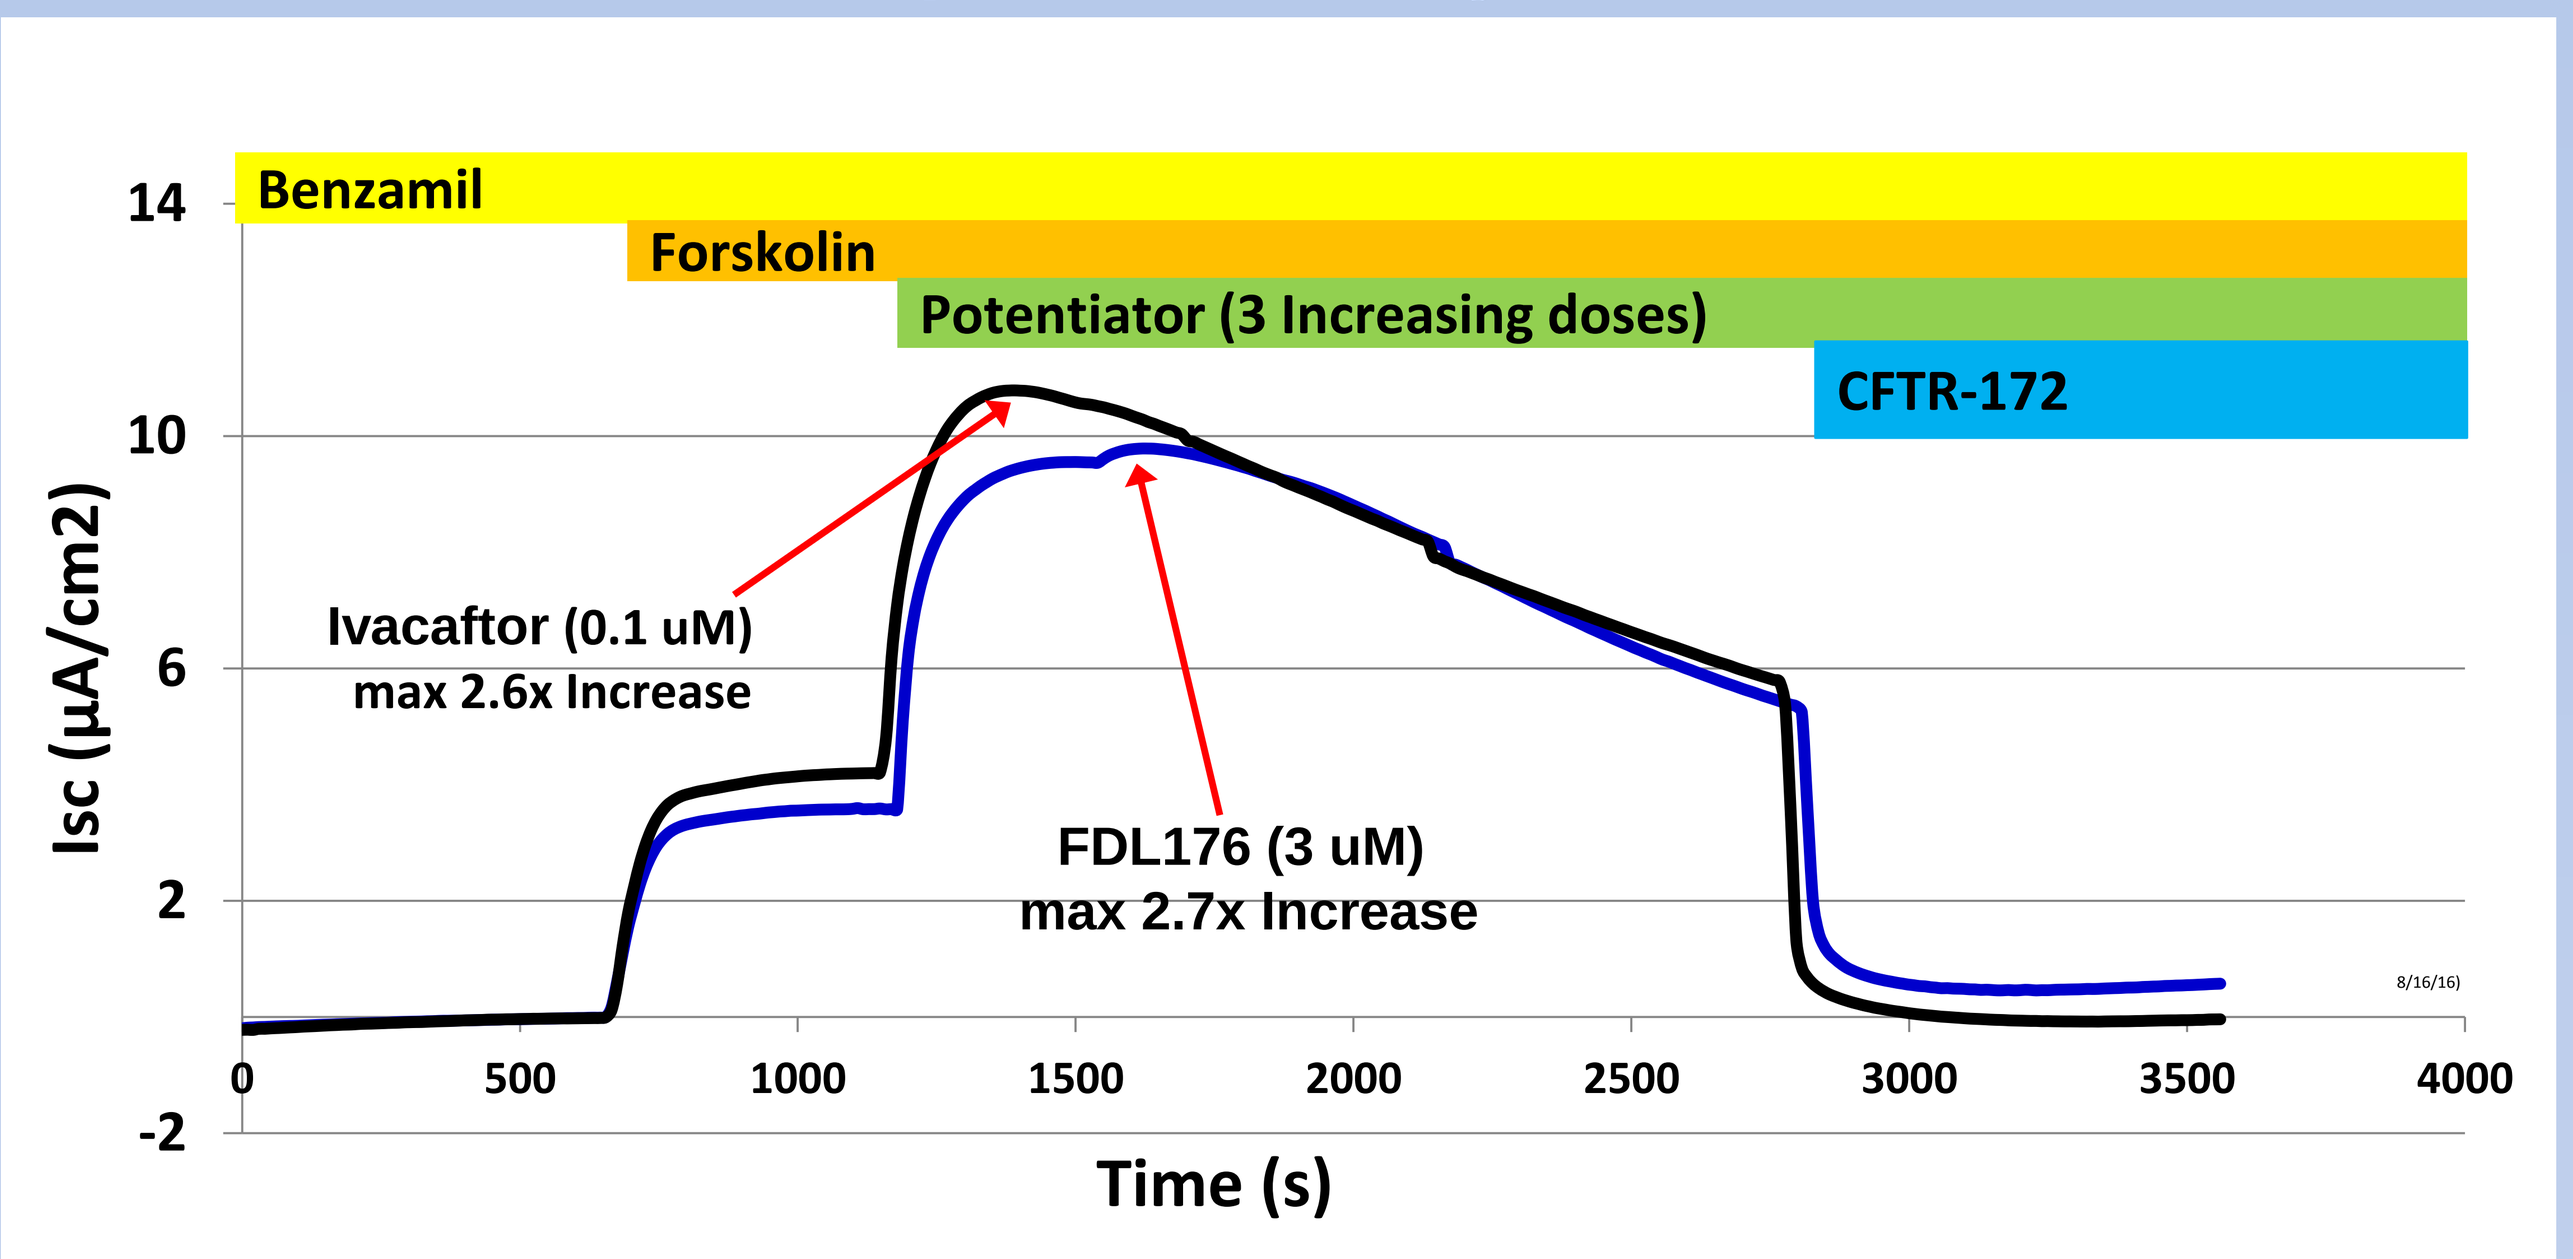
Benzamil
Forskolin
Potentiator (3 Increasing doses)
CFTR-172
Ivacaftor (0.1 uM)
max 2.6x Increase
FDL176 (3 uM)
max 2.7x Increase
Isc (µA/cm2)
Time (s)

- Data from 2 averaged traces of F508del-CFTR FRT cells in the Üssing Chamber (Isc)
- The FRT monolayers are exposed to a 15/1 chloride gradient which then applies a fixed known electrochemical driving force for Cl- to exit via CFTR
- V=IR (Ohm's Law) therefore I=V/R
- Isc assay measures I with constant V, therefore any change in current is due to an increased open probability of the F508del CFTR channel
- Stimulation with FDL176 increases chloride current >2-fold suggesting the open probability of uncorrected F508del-CFTR increases at least 2-fold

Figure 6: FDL176 Dose Response in G551D Mutation

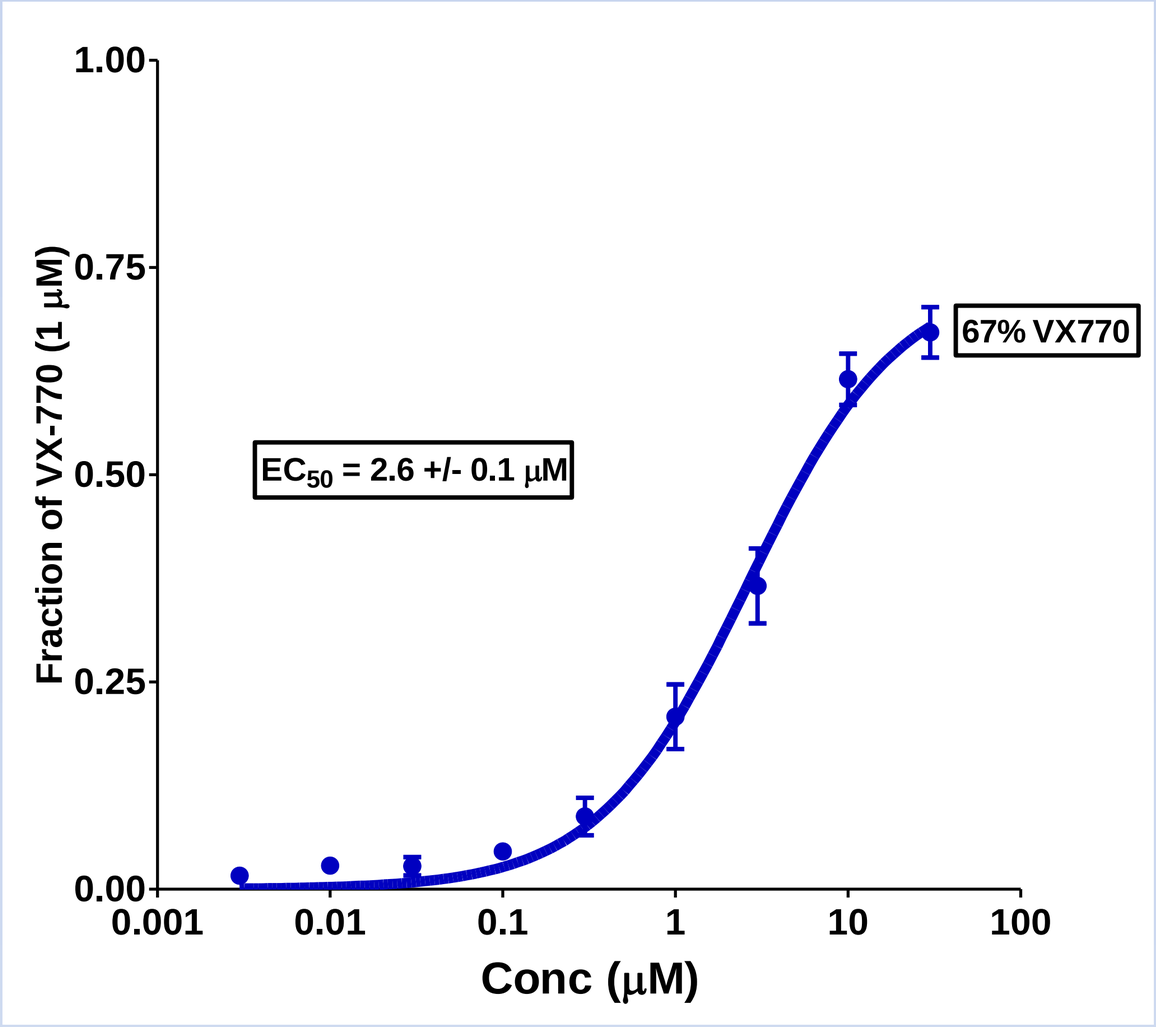
EC_{50} = 2.6 +/- 0.1 µM
67% VX770
Fraction of VX-770 (1 µM)
Conc (µM)

- Dose response curve of FDL176 in FRT cells expressing the G551D mutation
- Experiment conducted in the Üssing chamber (Isc assay) with a 15/1 chloride gradient
- Potency of FDL176 to potentiate G551D is higher compared to F508del-CFTR (also seen with Ivacaftor)
- The maximum efficacy of FDL176 in G551D FTR cells was observed at the top test concentration (30 µM)

Figure 7: FDL176 Potentiates CFTR with Various Mutations

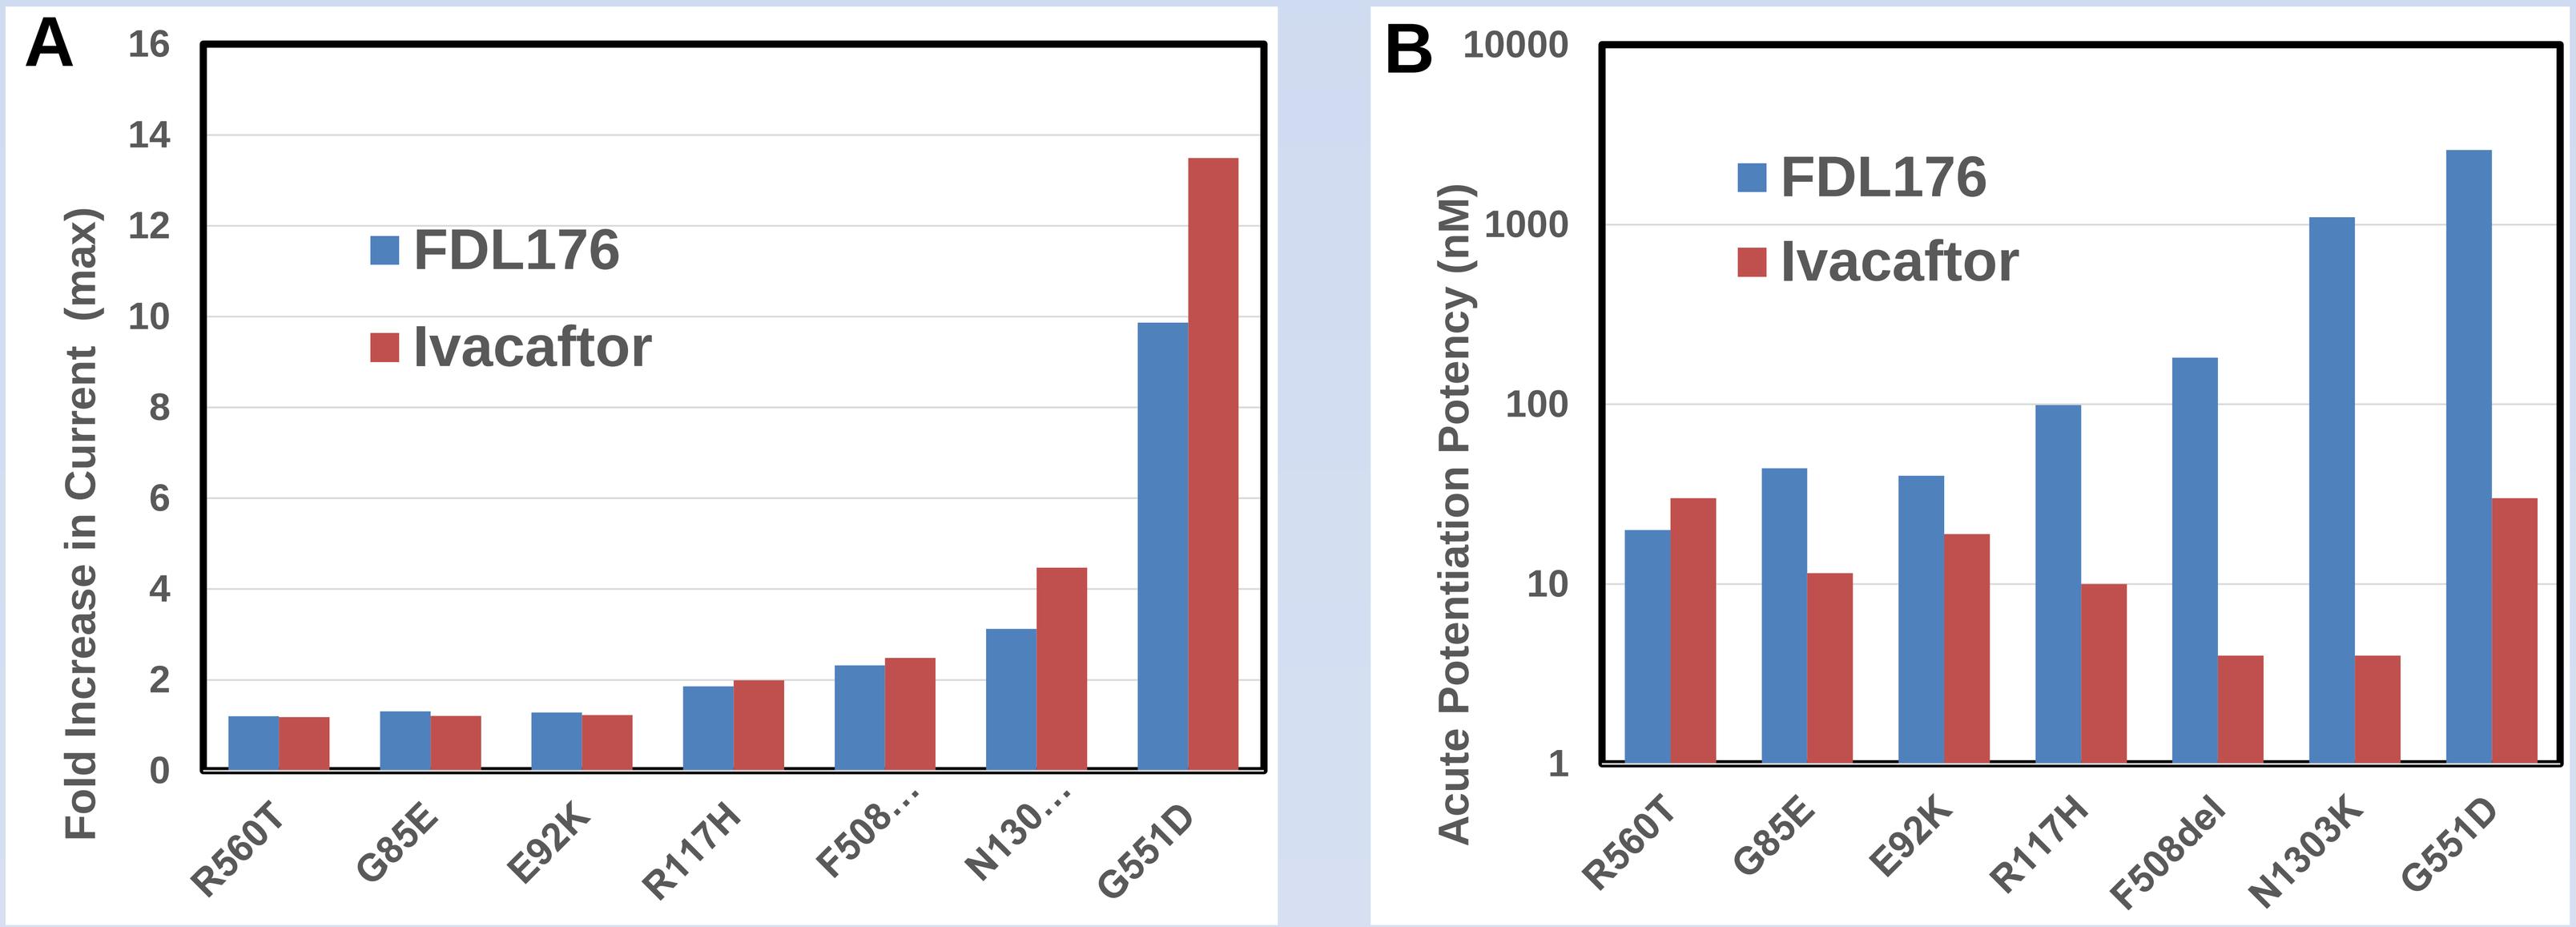
A
FDL176
Ivacaftor
Fold Increase in Current (max)
R560T
G85E
E92K
R117H
F508…
N130…
G551D
B
Acute Potentiation Potency (nM)
F508del
N1303K

- Figure A:Emax (fold increase over vehicle control); Figure B: Potency (EC_{50} nM)
- R560T, G85E, and E92K have almost no potentiation
- Gating mutations have the largest potentiation.

Summary

- FDL176 demonstrates similar maximum efficacy as ivacaftor in F508del-CFTR under acute potentiation conditions
- Chronic treatment of F508del-CFTR with FDL176 is more efficacious than chronic treatment with ivacaftor
- Human serum does not affect chronic inhibition but does increase CFTR current
- FDL176 increases open probability of F508del-CFTR
- FDL176 potentiates other CFTR mutations including G551D

Flatley Discovery Lab

Seeking A Cure for Cystic Fibrosis